196

Utilizing pharmacokinetic (PK) and pharmacodynamic (PD) biomarkers to support recommended Phase 2 dose (RP2D) selection for Phase 1 study of SAR444245 (SAR'245) in patients with advanced solid tumors

Siqing Fu[1], Meredith Mckean[2], Gerald Falchook[3], Minal Barve[4], Cheng Ean Chee[5], Charlotte Lemech[6], Tira J. Tan[7], Tarek Meniawy[8], Tatiana Hernández Guerrero[9], Wenting Wang[10], Adyb Baakili[11], Neyssa Marina[10†], Giovanni Abbadessa[10*], Silke Gastine[10], Rui Wang[10], Emiliano Calvo Aller[12]

*[1]Investigational Cancer Therapeutics, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, USA; [3]Sarah Cannon Research Institute, HealthONE, Denver, CO, USA; [4]Mary Crowley Cancer Research, Dallas, TX, USA; [5]Department of Hematology-Oncology, National University Cancer Institute, National University Health System, Singapore; [6]Scientia Clinical Research and Prince of Wales Clinical School, Randwick, NSW, Australia; [7]Division of Medical Oncology, National Cancer Centre, Singapore; [8]Linear Clinical Research & University of Western Australia, Nedlands, WA, Australia; [9]START Barcelona-HM Nou Delfos, Barcelona, Spain; [10]Sanofi, Cambridge, MA, USA; [11]Sanofi, Paris, France; [12]START Madrid-CIOCC, Centro Integral Oncológico Clara Campal, Madrid, Spain. †Current affiliation: Arcus Biosciences; *Current affiliation: ModeX Therapeutics Inc.*

BACKGROUND

- SAR444245 (SAR'245) is a clinical-stage, site-specific PEGylated human non-alpha interleukin-2 (IL-2) that selectively engages IL-2 alpha receptor binding but retains near-native–binding affinity for beta/gamma complex[1]
- This results in a unique 'T-cell remodeling' mechanism of action (MoA), characterized by robust increase in $CD8^+$ T cells, coupled with potent natural killer (NK) cell activation/expansion without inducing regulatory T-cell expansion (T_{reg})[2]
- HAMMER is a Phase 1/2, first-in-human (FIH), open label, multicenter, dose escalation and dose expansion study of SAR'245 as a single agent and as a combination therapy in participants with advanced or metastatic solid tumors[3]
- The selection of dose and dosing schedule for therapies targeting interleukin-2 (IL-2) and IL-2 receptor (IL-2R) is complex due to the careful balance between maximizing therapeutic efficacy and minimizing toxicity[4]
- SAR'245 was dosed at less frequent dosing schedule, at every 3 weeks (Q3W) or every 2 weeks (Q2W). More intensive dosing schedule every week (QW) was explored, after the failure of other well-known engineered IL-2R agonist, to induce more robust MoA biomarker modulation[5,6]
- Herein, we integrate various clinical parameters, including pharmacokinetic (PK), pharmacodynamic (PD) biomarkers, and safety, to contribute to the selection of recommended Phase 2 dose (RP2D) for SAR'245

METHODS

- Intravenous SAR'245 monotherapy was administered Q3W (Cohort B), Q2W (Cohort A), or QW (Cohort G) (**Figure 1**)
- Peripheral blood was collected for immune cell profiling, including circulating $CD8^+$ T cell, NK cell, and T_{reg} cells
- Efficacy surrogate biomarker circulating DNA (ctDNA) was measured from serum by Guardant Health
- A semi-mechanistic population PK/PD model was developed to depict IL-2–induced cell trafficking away from blood to expansion sites immediately after administration and subsequent reappearance of expanded cells in blood
- RP2D selected based on the totality of safety, PK/PD association, and efficacy surrogate biomarker data will be reported elsewhere as a part of the clinical analysis

Study design

Figure 1: Phase 1/2 HAMMER study design (monotherapy cohorts A, B, and G)

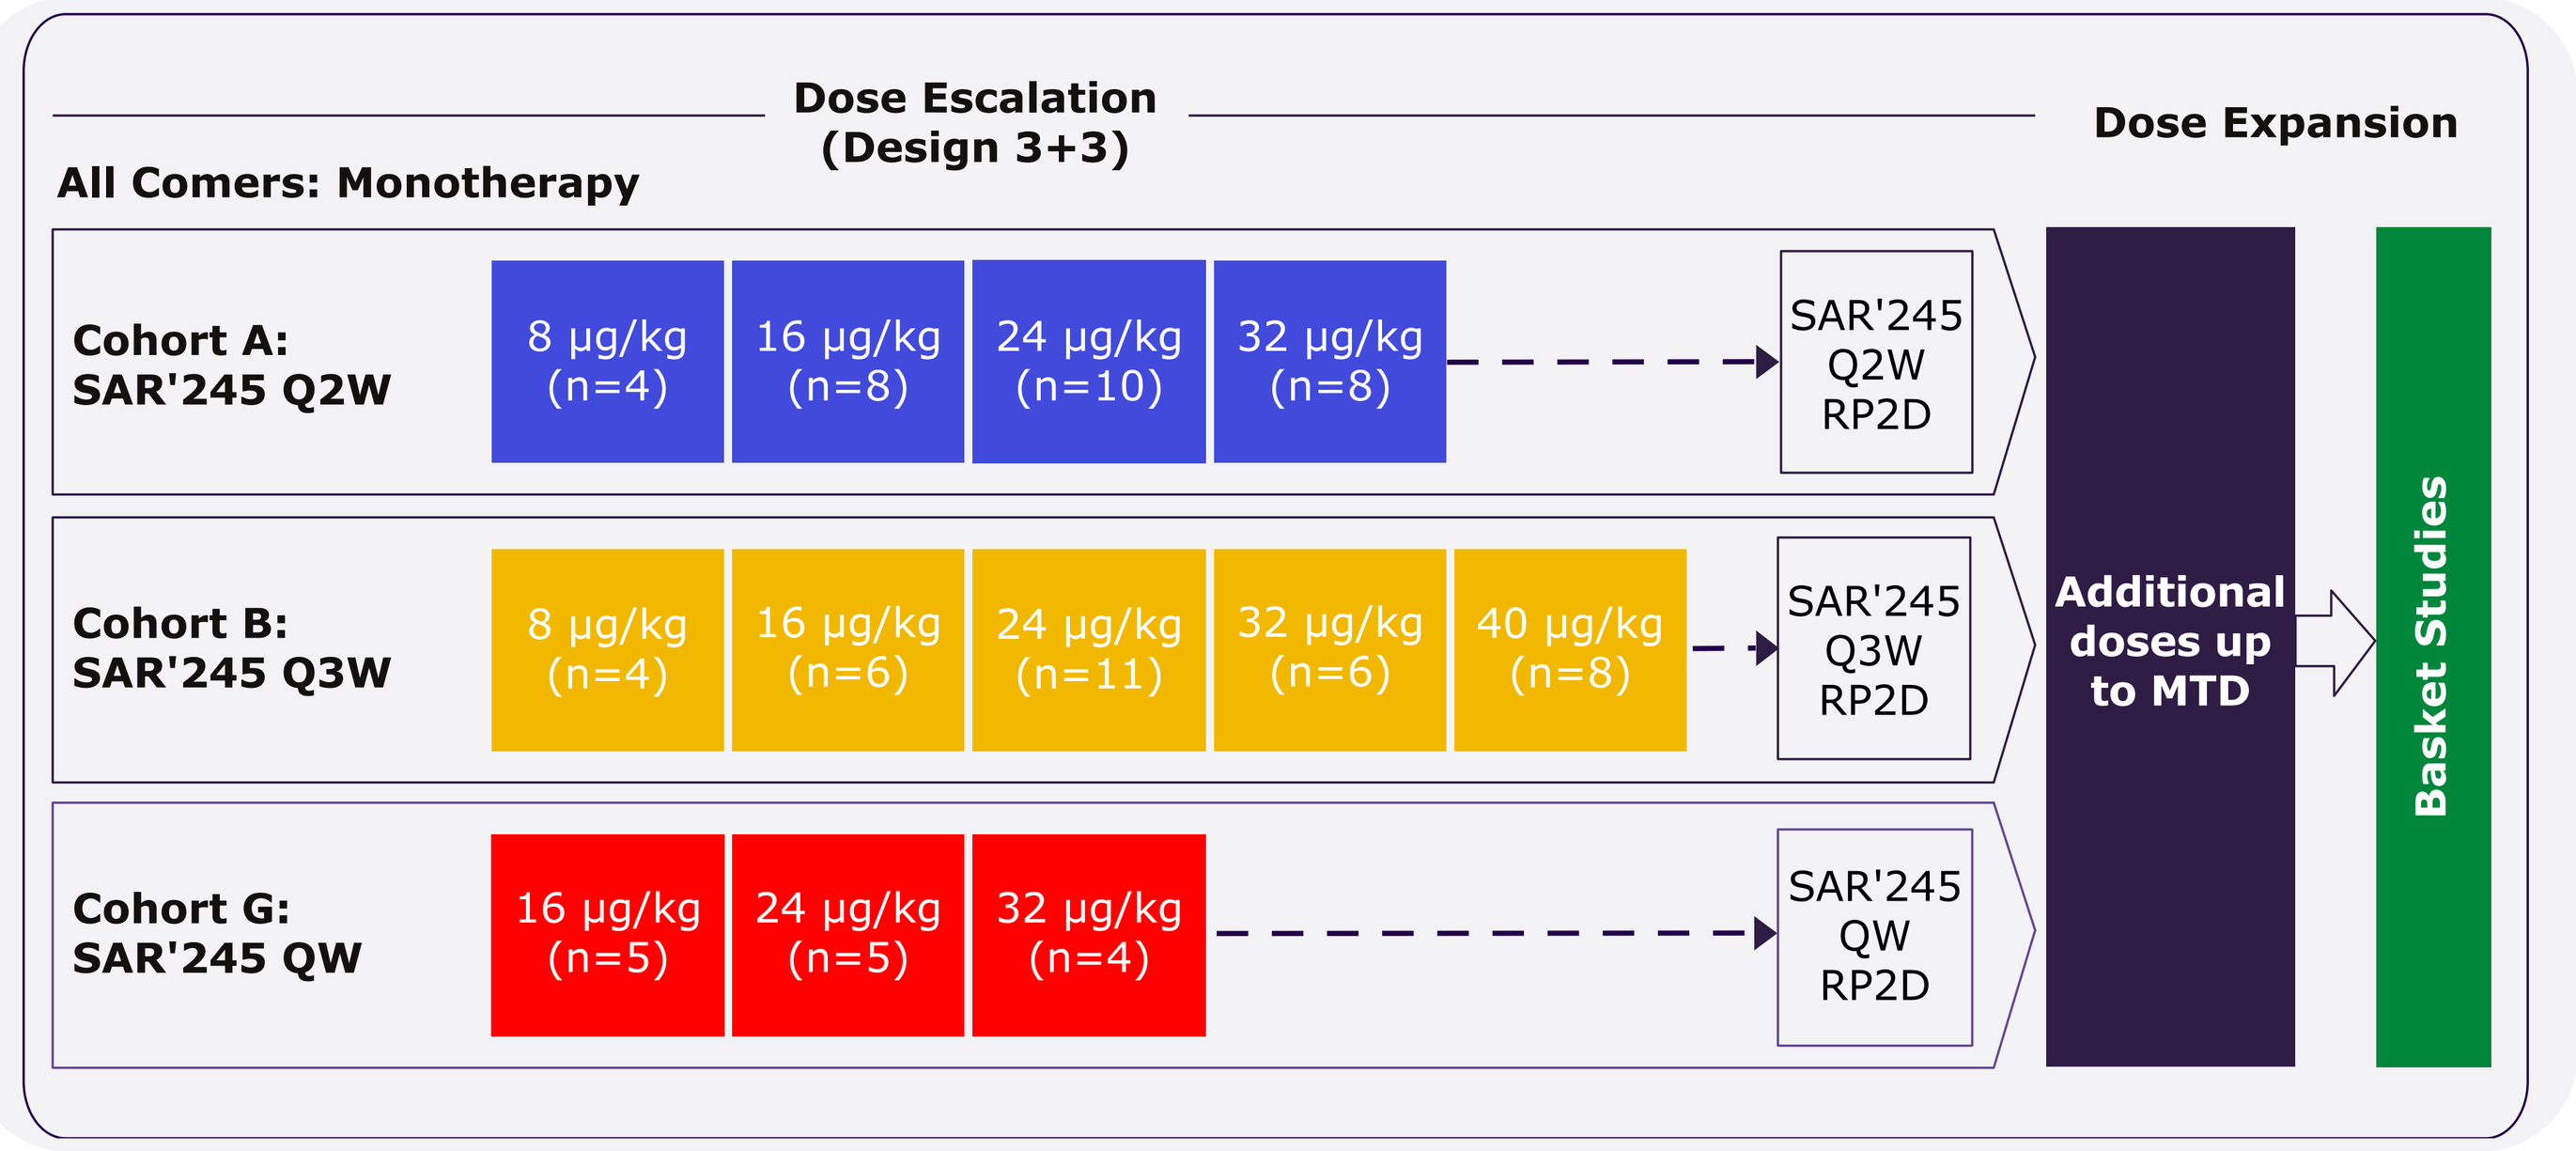
Dose Escalation
(Design 3+3)
Dose Expansion
All Comers: Monotherapy
Cohort A:
SAR'245 Q2W
8 µg/kg (n=4)
16 µg/kg (n=8)
24 µg/kg (n=10)
32 µg/kg (n=8)
SAR'245 Q2W RP2D
Cohort B:
SAR'245 Q3W
8 µg/kg (n=4)
16 µg/kg (n=6)
24 µg/kg (n=11)
32 µg/kg (n=6)
40 µg/kg (n=8)
SAR'245 Q3W RP2D
Cohort G:
SAR'245 QW
16 µg/kg (n=5)
24 µg/kg (n=5)
32 µg/kg (n=4)
SAR'245 QW RP2D
Additional doses up to MTD
Basket Studies

MTD, maximum tolerated dose; QW, every week; Q2W, every 2 weeks; Q3W, every 3 weeks; RP2D, recommended Phase 2 dose; SAR'245, SAR444245.

RESULTS

- As of September 2024, samples from 30 (Cohort A), 35 (Cohort B), and 14 (Cohort G) subjects were available

Comparing lymphocyte expansion induced at various dosing schedules

- Dose-dependent peak fold change of lymphocyte expansion was observed in Cohort G: 16 vs. 24 vs. 32 µg/kg (**Figure 2**)
- QW dosing schedule of SAR'245 at 16, 24, and 32 µg/kg, results in greater lymphocyte expansion compared to Q2W dosing schedule at the same dose levels (**Figure 2**)

Figure 2: Peak fold change (mean±SEM) of lymphocyte expansion observed during cycle 1 or cycle 2 compared to baseline

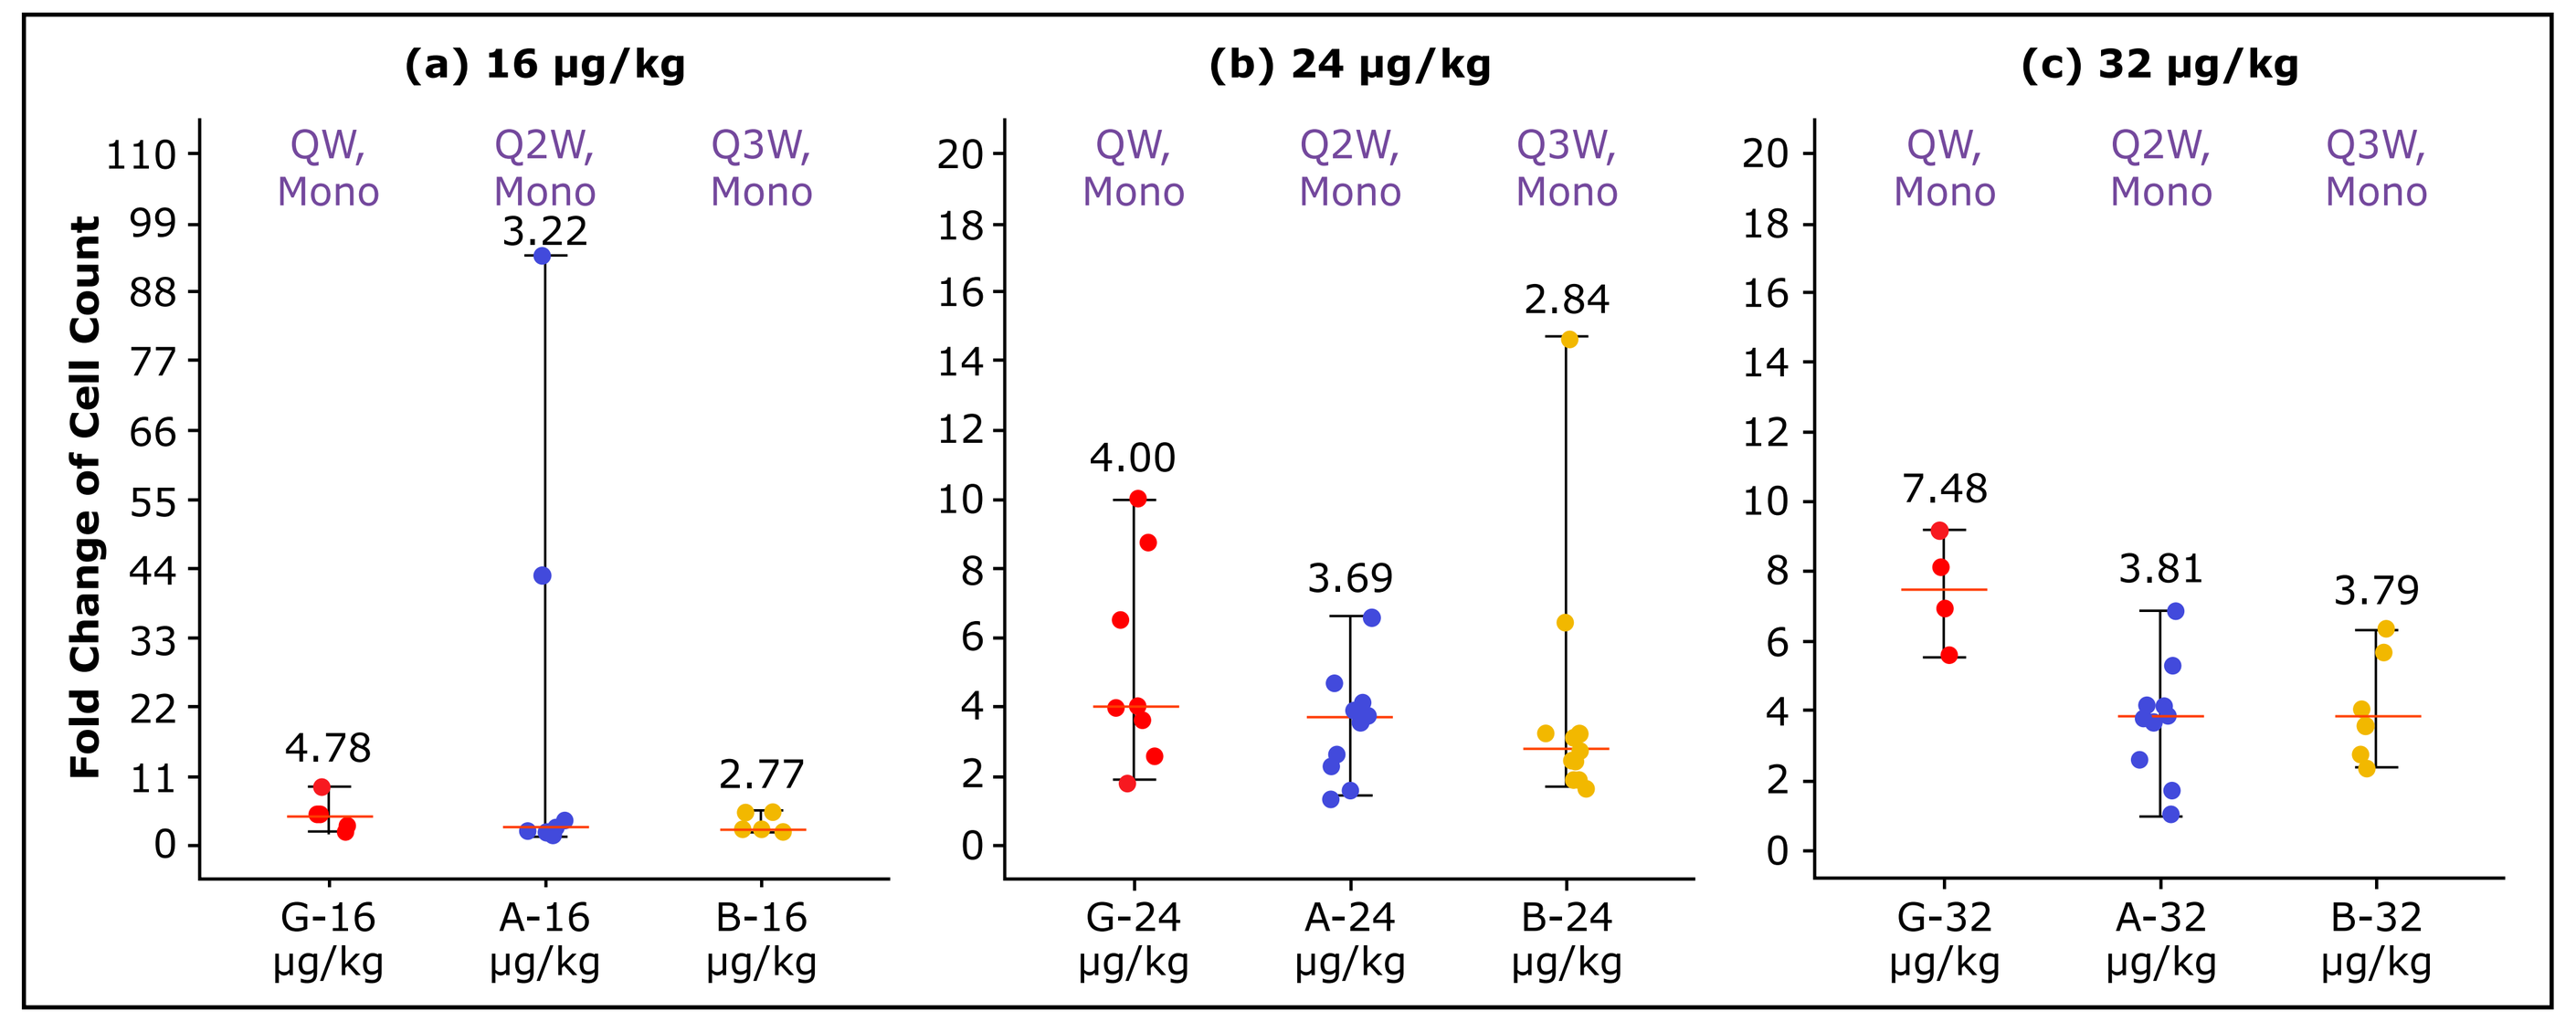
(a) 16 µg/kg
(b) 24 µg/kg
(c) 32 µg/kg
Fold Change of Cell Count
QW, Mono
Q2W, Mono
Q3W, Mono
4.78
3.22
2.77
4.00
3.69
2.84
7.48
3.81
3.79
G-16 µg/kg
A-16 µg/kg
B-16 µg/kg
G-24 µg/kg
A-24 µg/kg
B-24 µg/kg
G-32 µg/kg
A-32 µg/kg
B-32 µg/kg

Mono, monotherapy; QW, every week; Q2W, every 2 weeks; Q3W, every 3 weeks; SEM, standard error of mean.

Comparing $CD8^+$ T cell expansion induced at various dosing schedules

- At 16 µg/kg, more $CD8^+$ T cell expansion was induced by QW>Q2W>Q3W **(Figure 3a)**
- At 24 µg/kg and 32 µg/kg, the $CD8^+$ T cell expansion were similar among different dosing schedules **(Figure 3b and 3c)**

Figure 3: Peak fold change (mean±SEM) of $CD8^+$ T cell observed during cycle 1 or cycle 2 compared to baseline

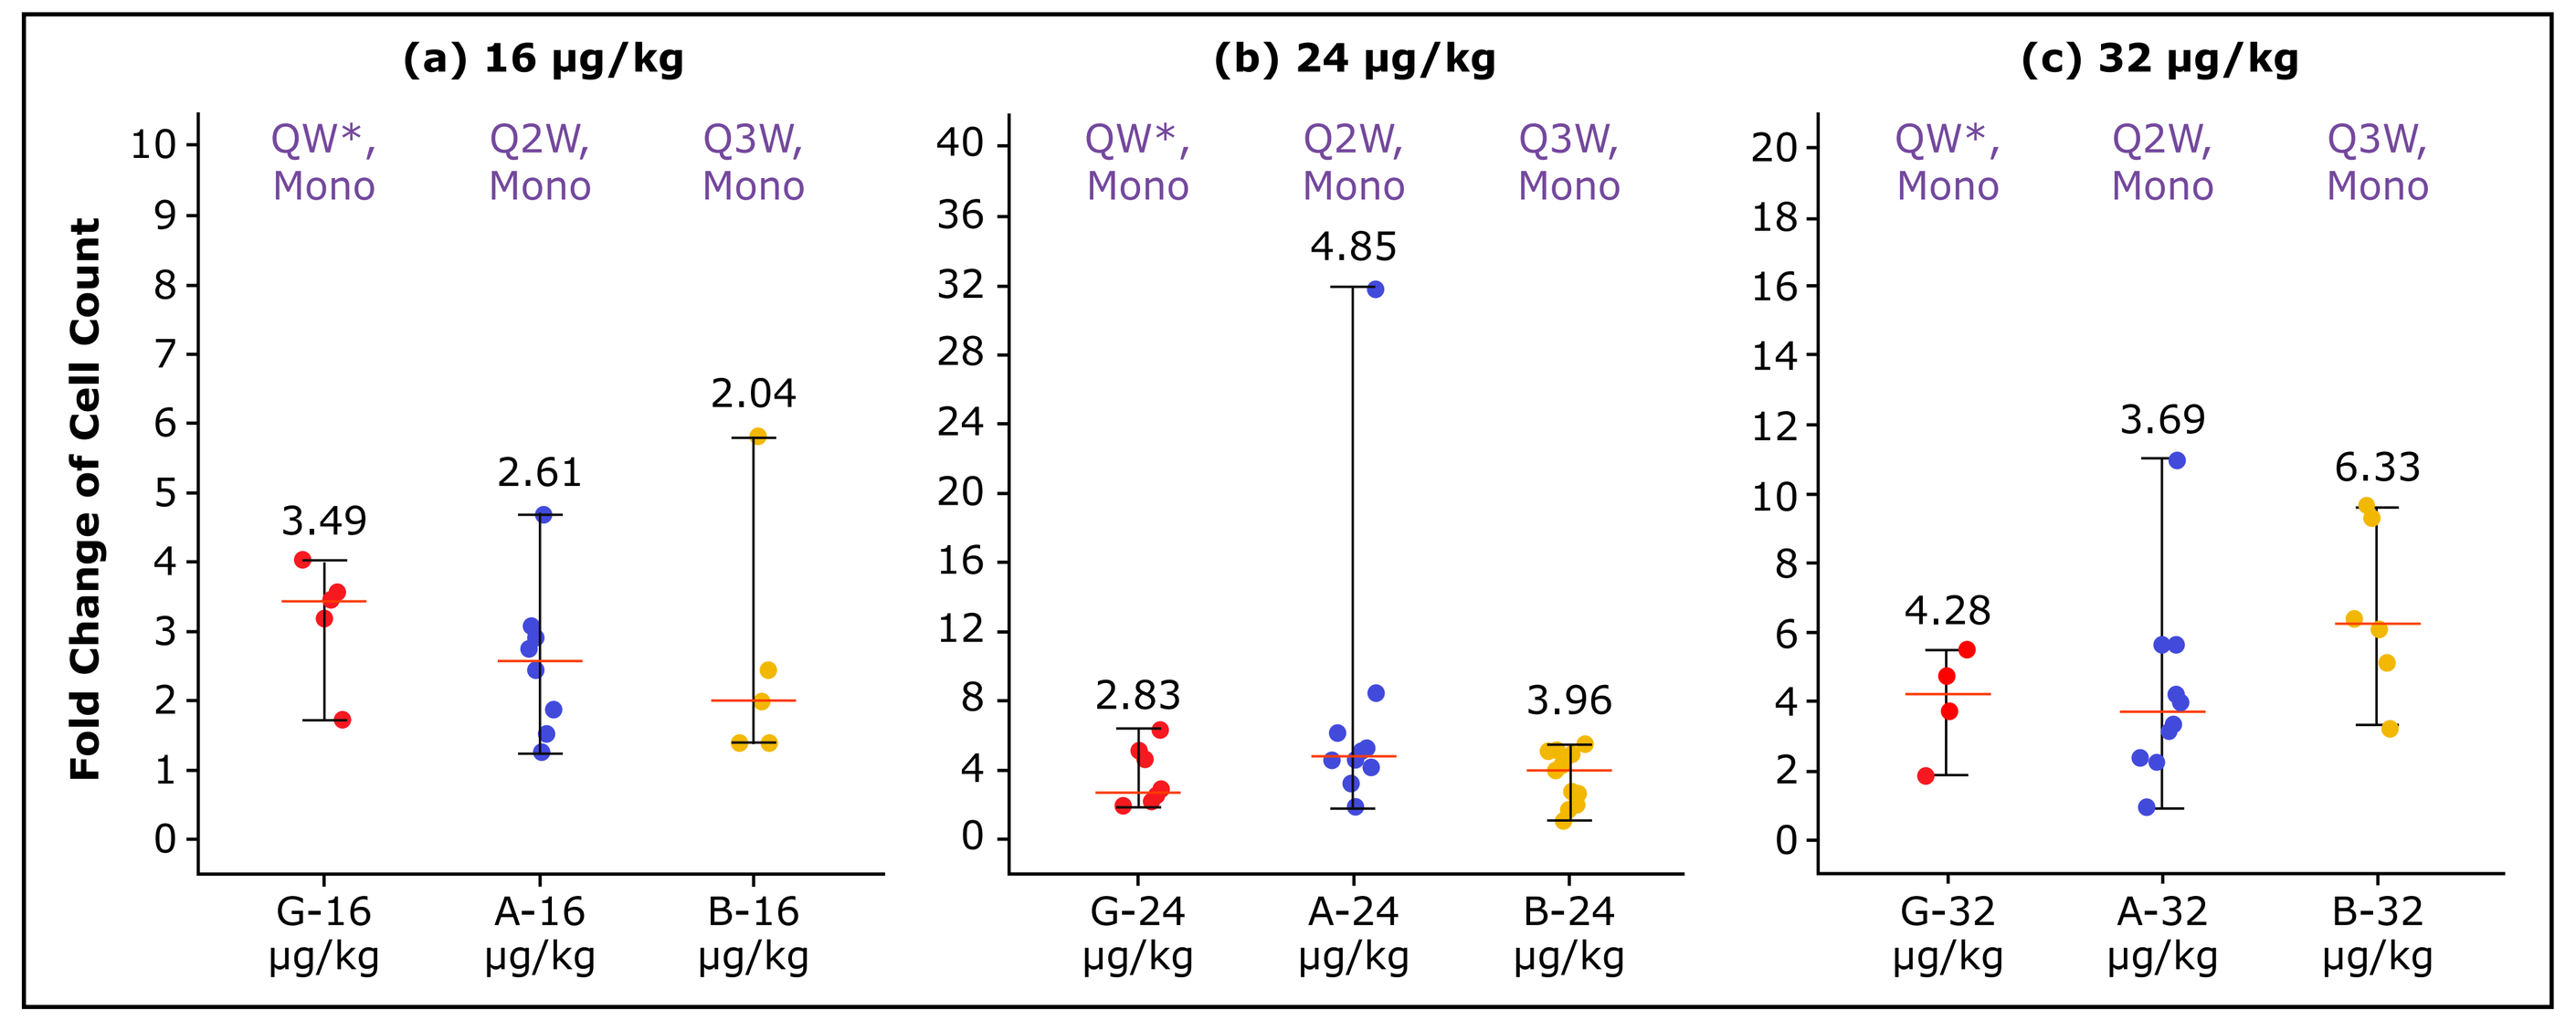
(a) 16 µg/kg
(b) 24 µg/kg
(c) 32 µg/kg
Fold Change of Cell Count
QW*, Mono
Q2W, Mono
Q3W, Mono
3.49
2.61
2.04
2.83
4.85
3.96
4.28
3.69
6.33
G-16 µg/kg
A-16 µg/kg
B-16 µg/kg
G-24 µg/kg
A-24 µg/kg
B-24 µg/kg
G-32 µg/kg
A-32 µg/kg
B-32 µg/kg

*Sample size of Cohort G was smaller as compared to Cohorts A and B.
CD, cluster of differentiation; Mono, monotherapy; QW, every week; Q2W, every 2 weeks; Q3W, every 3 weeks.

Comparing NK cell expansion induced at various dosing schedules

- At 16 µg/kg, more NK cell expansion was induced by QW>Q2W>Q3W **(Figure 4a)**
- At 24 µg/kg and 32 µg/kg, the NK cell expansion were similar among different dosing schedules **(Figure 4b and 4c)**

Figure 4: Peak fold change (mean±SEM) of NK cell observed during cycle 1 or cycle 2 compared to baseline

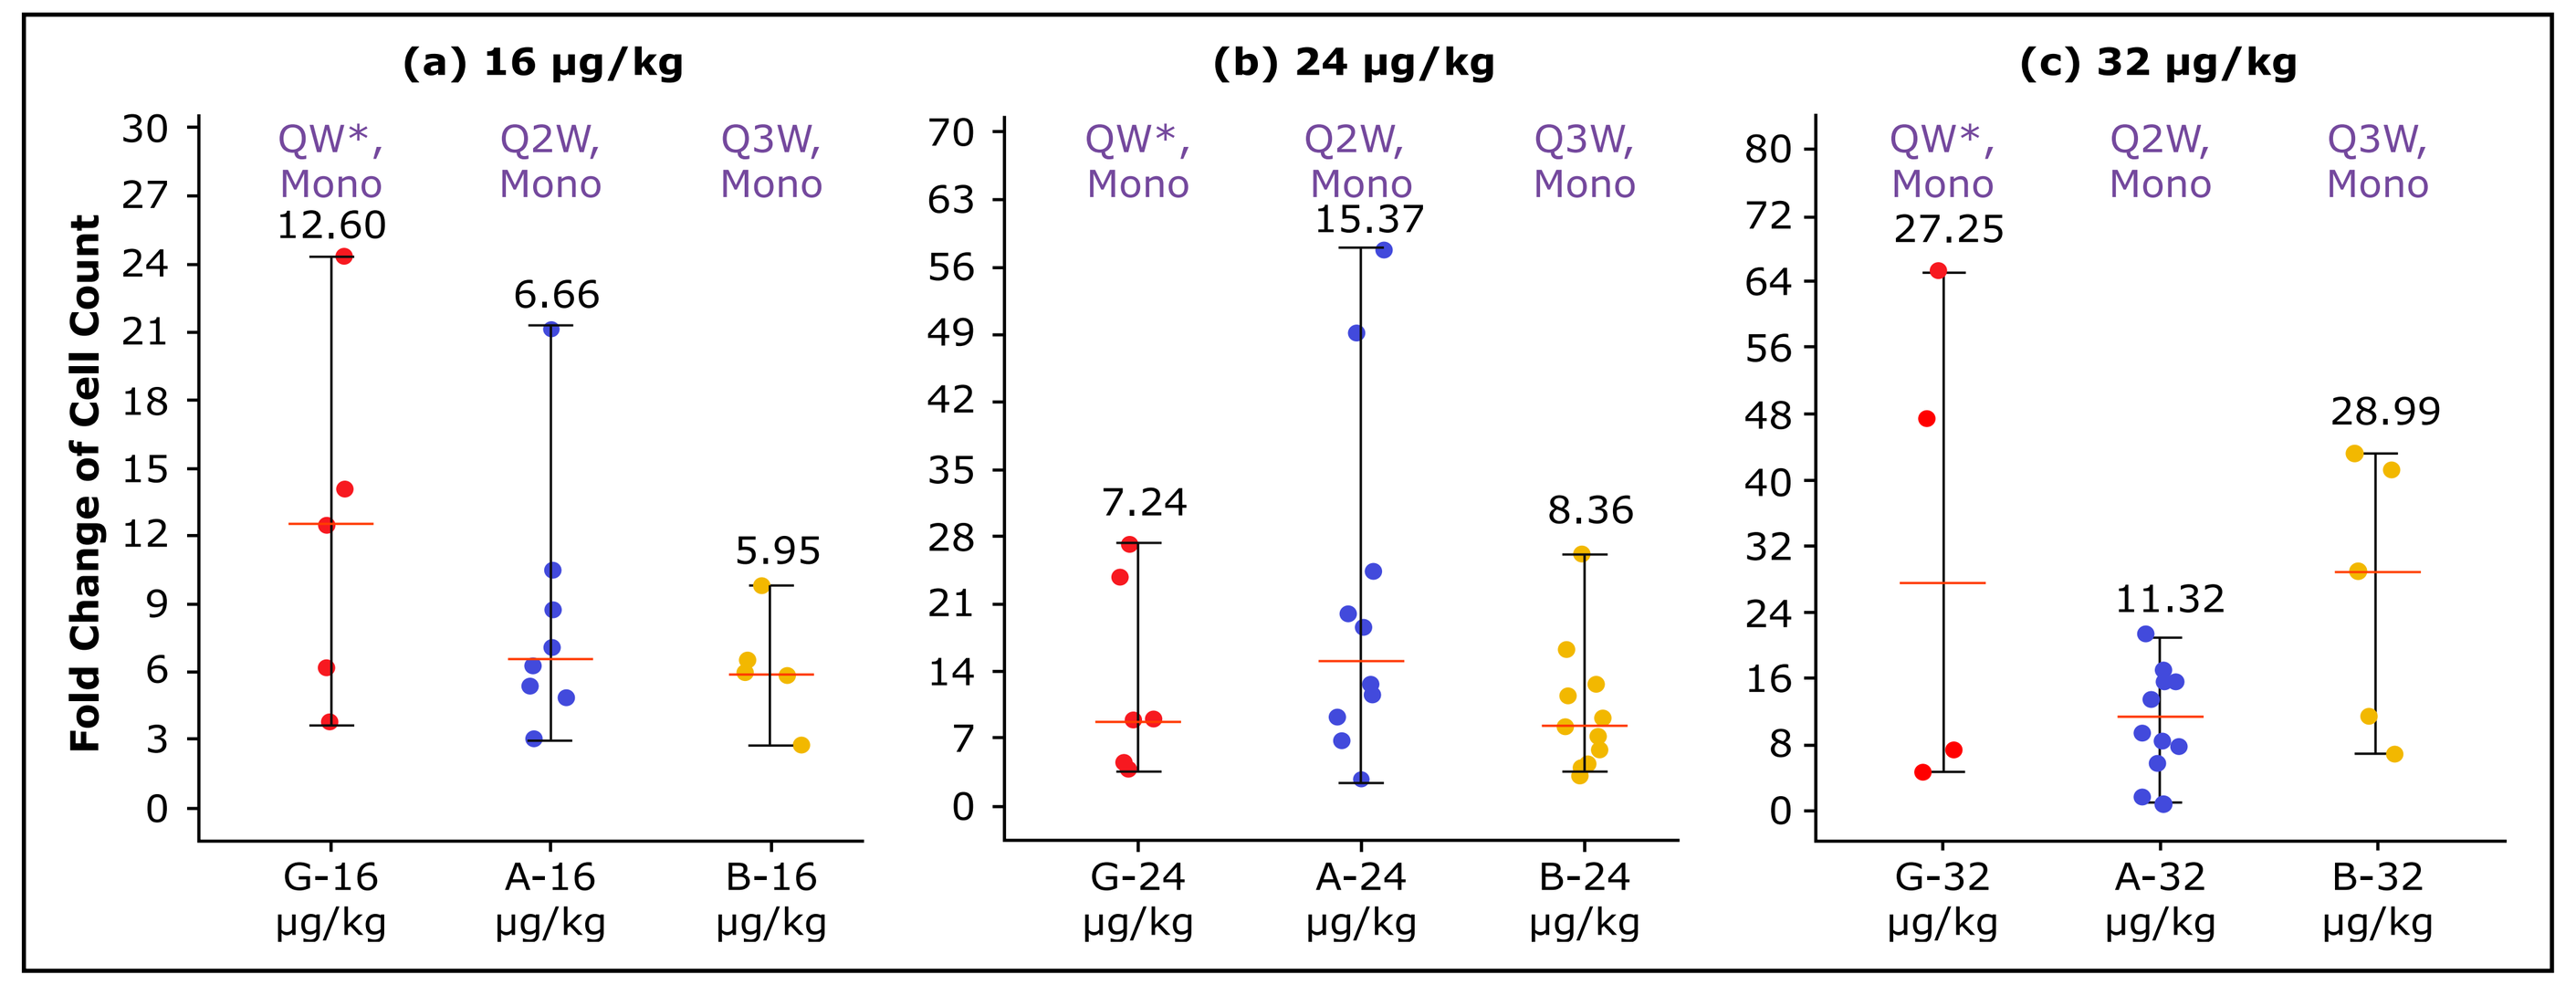
(a) 16 µg/kg
(b) 24 µg/kg
(c) 32 µg/kg
Fold Change of Cell Count
QW*, Mono
Q2W, Mono
Q3W, Mono
12.60
6.66
5.95
7.24
15.37
8.36
27.25
11.32
28.99
G-16 µg/kg
A-16 µg/kg
B-16 µg/kg
G-24 µg/kg
A-24 µg/kg
B-24 µg/kg
G-32 µg/kg
A-32 µg/kg
B-32 µg/kg

*Sample size of Cohort G was smaller as compared to Cohorts A and B.
NK, natural killer; Mono, monotherapy; QW, every week; Q2W, every 2 weeks; Q3W, every 3 weeks.

Flat modulation of peripheral $CD4^+$ T_{reg} across all dosing schedules

- There was no increased T_{reg} modulation when compared to Q2W and Q3W monotherapy **(Figure 5)**

Figure 5: Peak fold change (mean±SEM) of T_{reg} observed during cycle 1 or cycle 2 compared to baseline

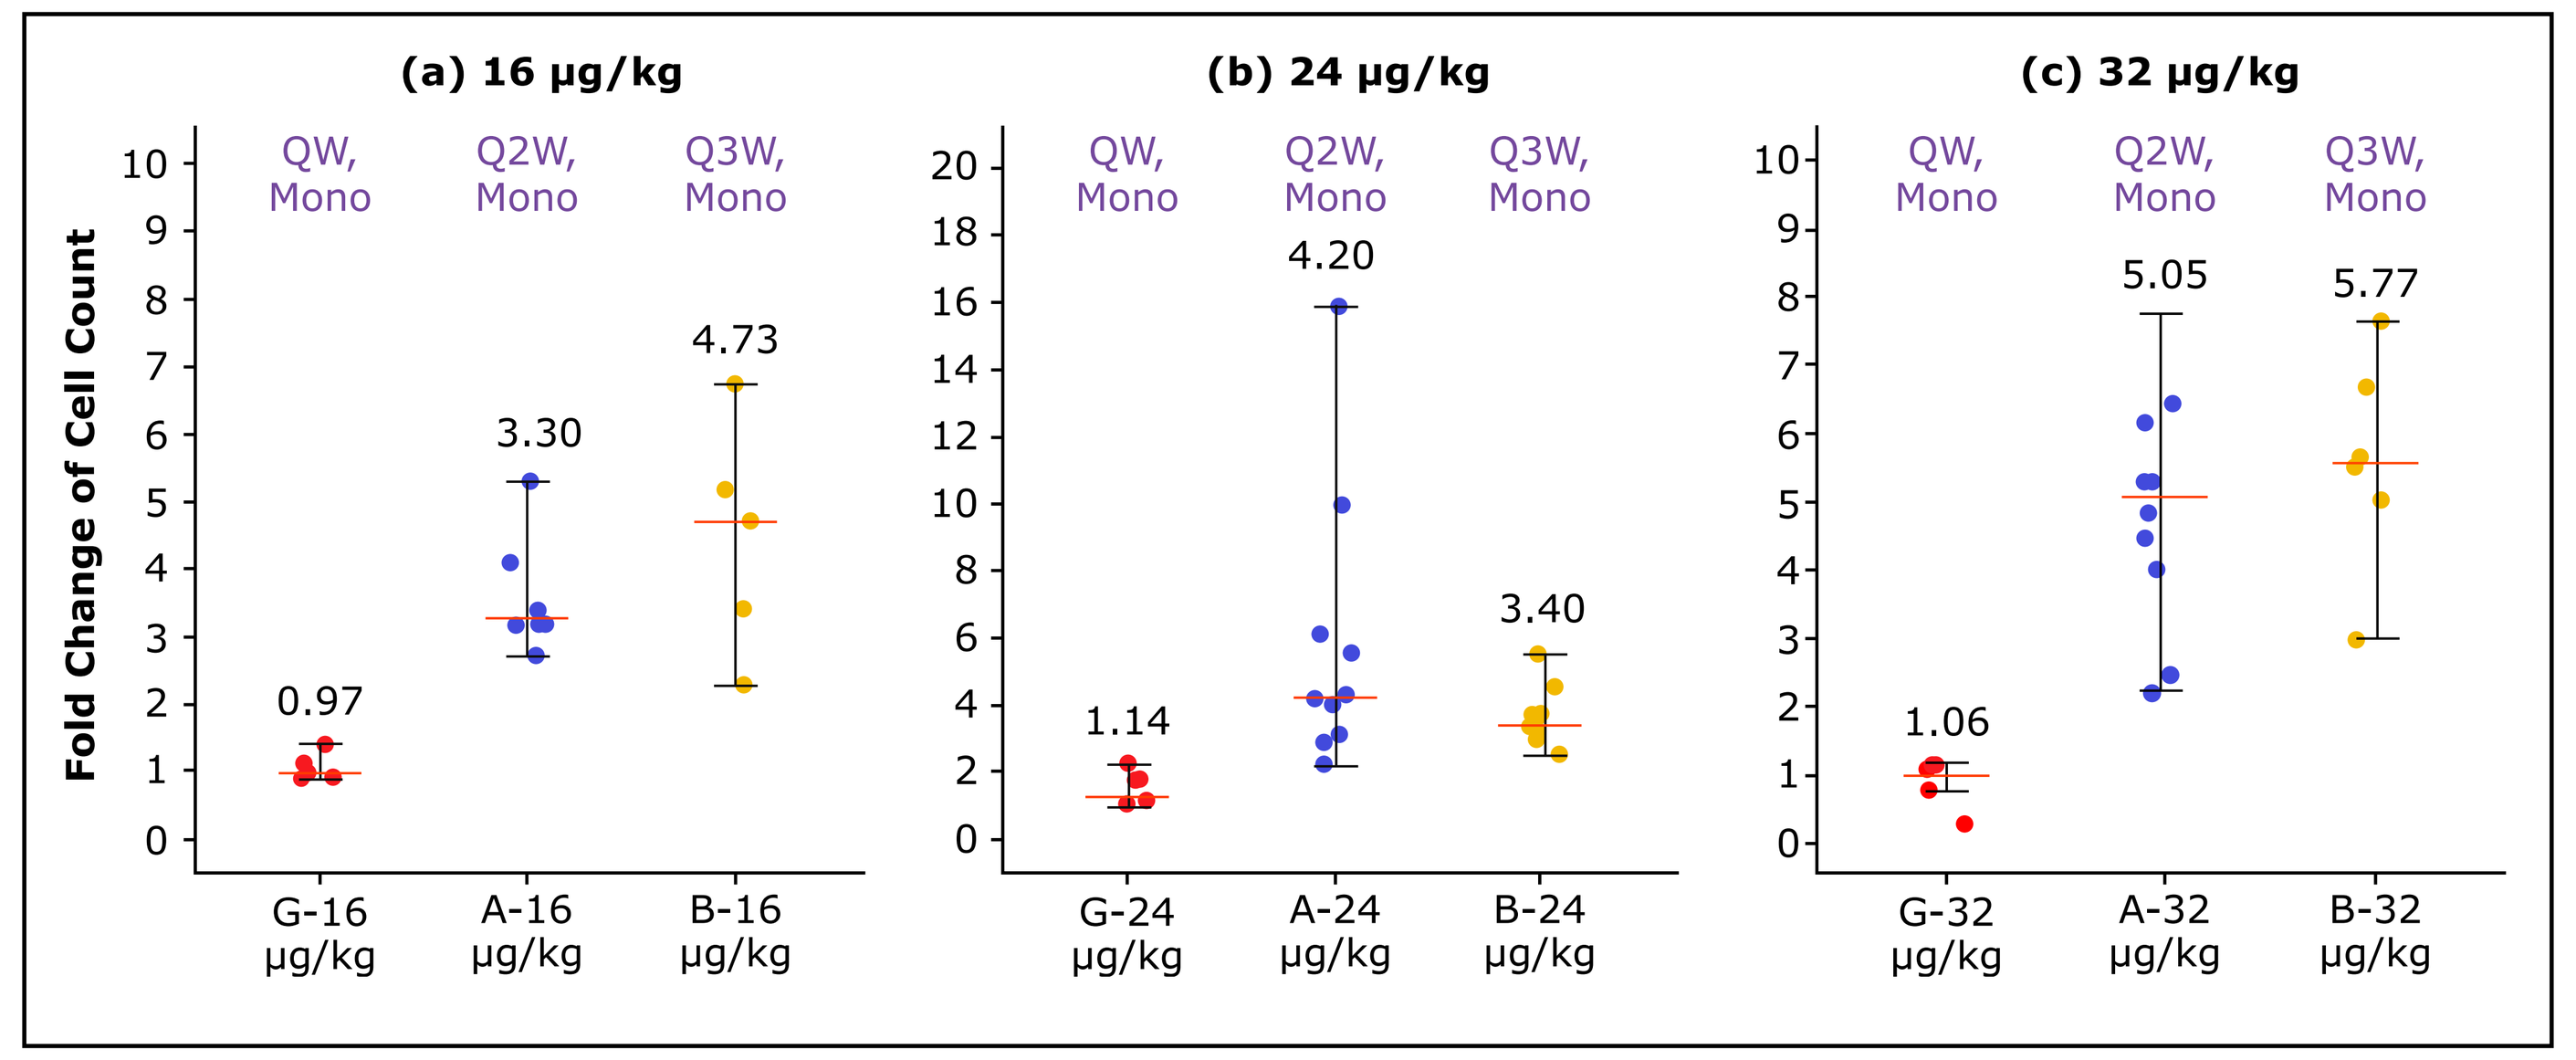
(a) 16 µg/kg
(b) 24 µg/kg
(c) 32 µg/kg
Fold Change of Cell Count
QW, Mono
Q2W, Mono
Q3W, Mono
0.97
3.30
4.73
1.14
4.20
3.40
1.06
5.05
5.77
G-16 µg/kg
A-16 µg/kg
B-16 µg/kg
G-24 µg/kg
A-24 µg/kg
B-24 µg/kg
G-32 µg/kg
A-32 µg/kg
B-32 µg/kg

CD, cluster of differentiation; Mono, monotherapy; QW, every week; Q2W, every 2 weeks; Q3W, every 3 weeks; T_{reg}, regulatory T cell.

Comparing $CD8^+$ T cell expansion induced at various dosing schedules

- QW dosing induced more sustainable $CD8^+$ T cell expansion
- We start observing a plateauing effect from 24 to 32 µg/kg **(Figure 6)**

Figure 6: Peak fold change of $CD8^+$ T cell expansion across QW, Q2W and Q3W (simulated data)

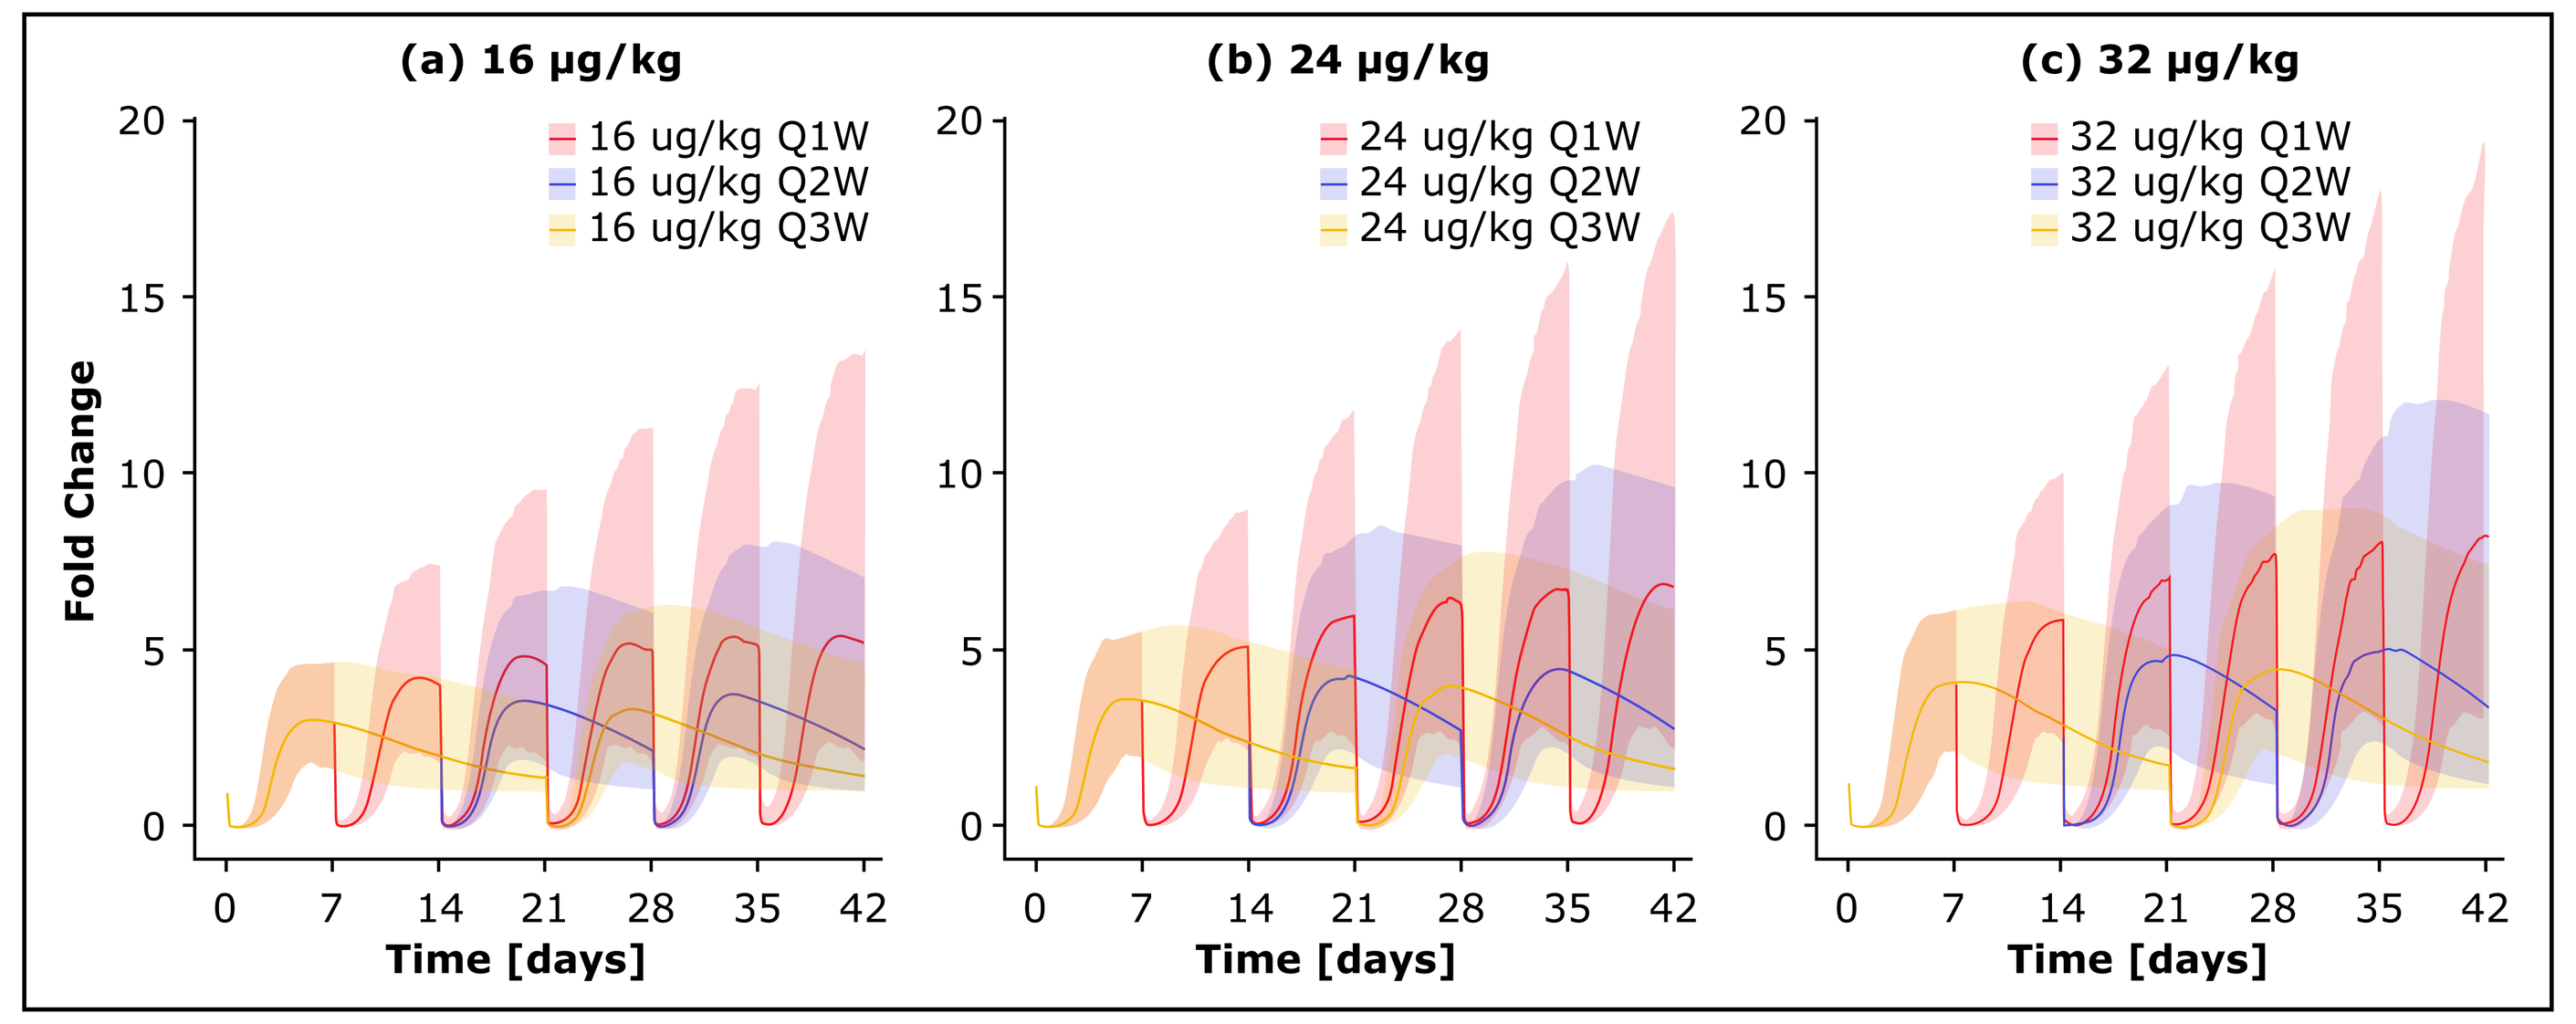
(a) 16 µg/kg
(b) 24 µg/kg
(c) 32 µg/kg
16 ug/kg Q1W
16 ug/kg Q2W
16 ug/kg Q3W
24 ug/kg Q1W
24 ug/kg Q2W
24 ug/kg Q3W
32 ug/kg Q1W
32 ug/kg Q2W
32 ug/kg Q3W
Fold Change
Time [days]

CD, cluster of differentiation; QW, every week; Q2W, every 2 weeks; Q3W, every 3 weeks.

Comparing NK expansion induced at various dosing schedules

- QW dosing induced more sustainable NK expansion
- We start observing a plateauing effect comparing 24 µg/kg to 16 µg/kg **(Figure 7)**

Figure 7: Peak fold change of NK cell expansion across QW, Q2W and Q3W (simulated data)

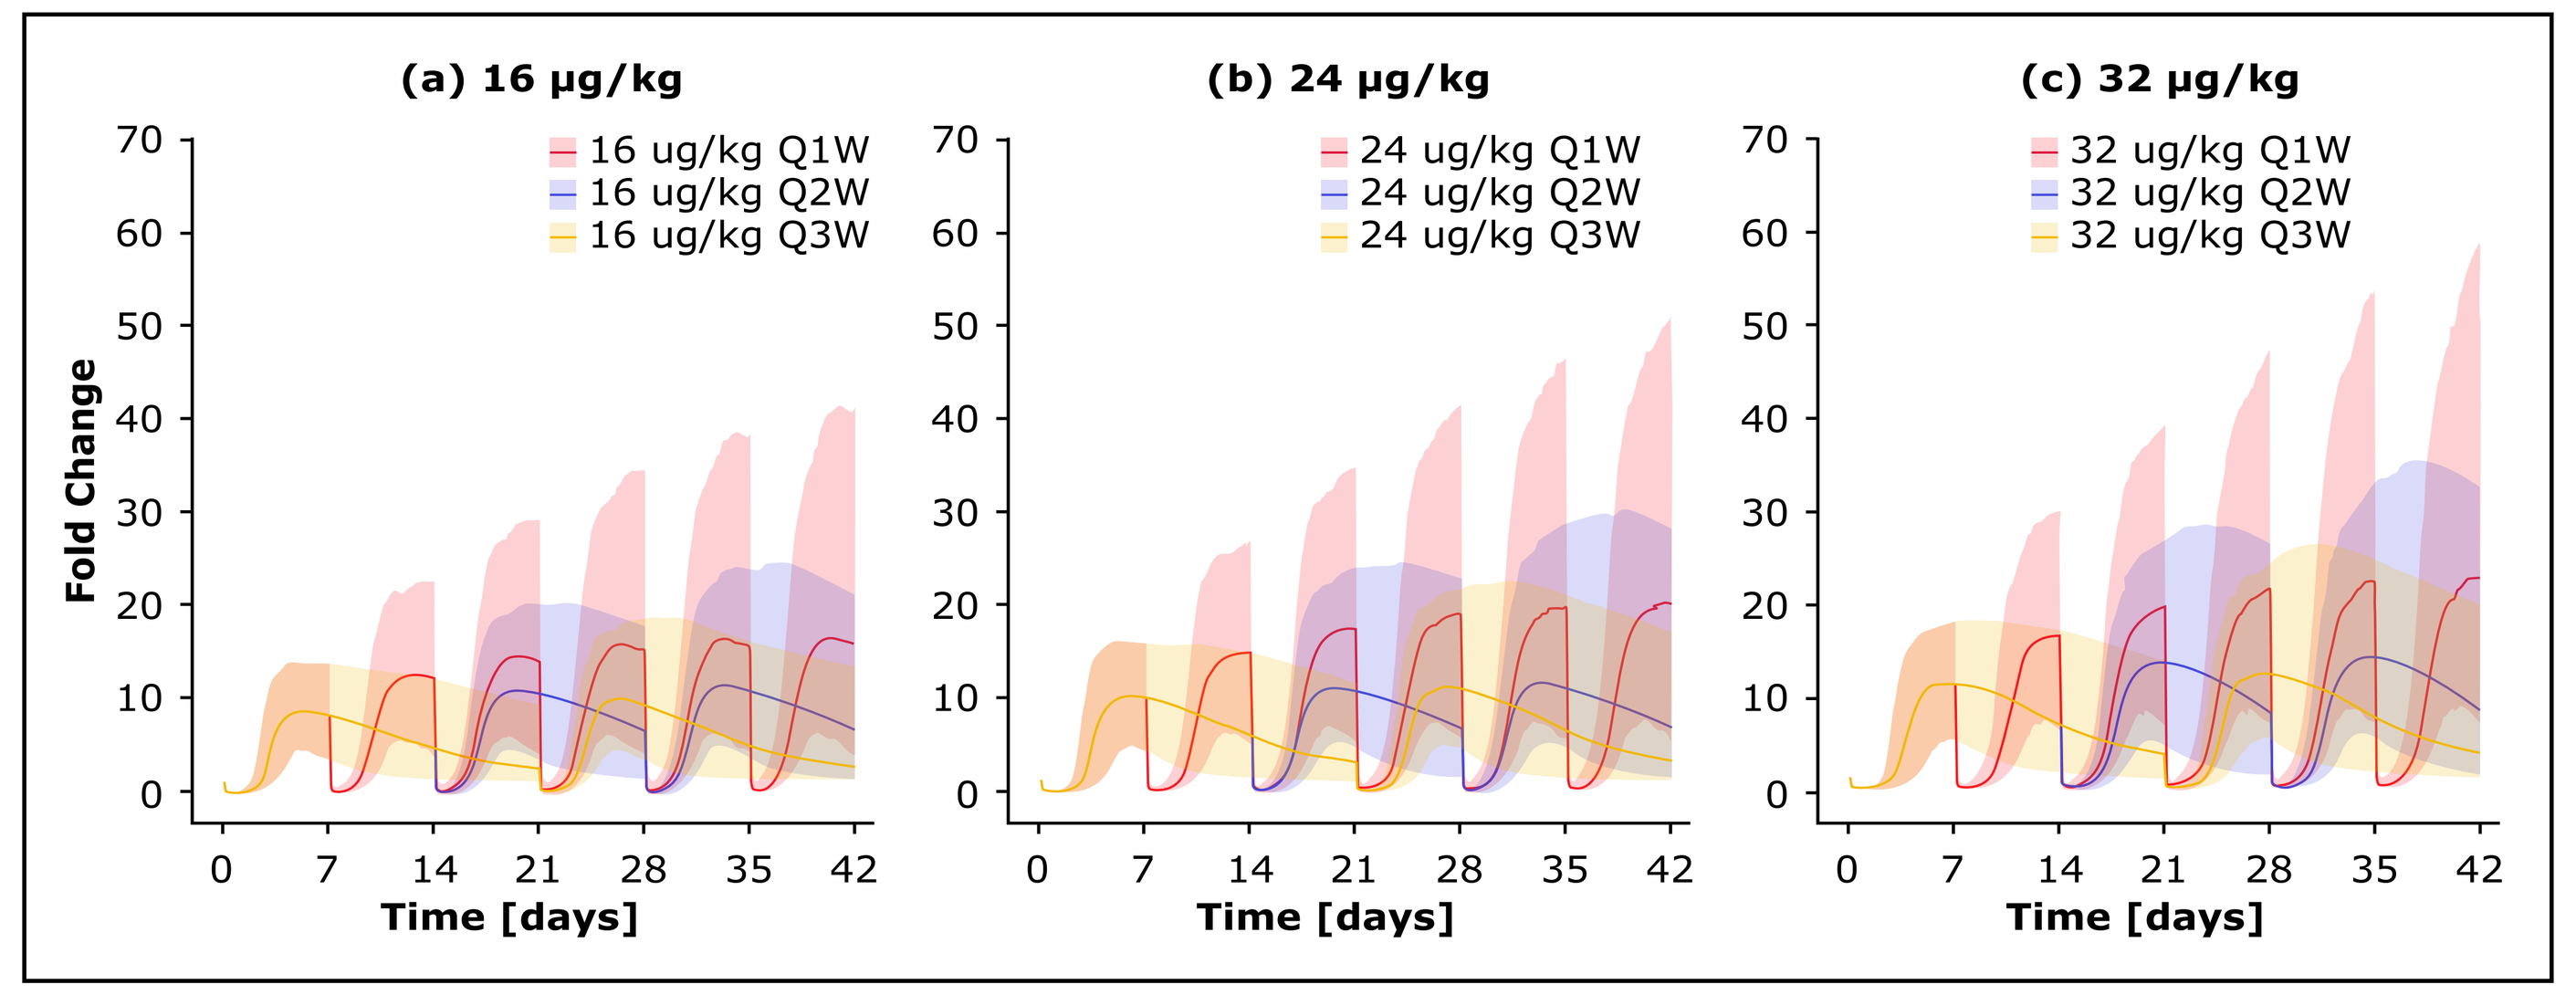
(a) 16 µg/kg
(b) 24 µg/kg
(c) 32 µg/kg
16 ug/kg Q1W
16 ug/kg Q2W
16 ug/kg Q3W
24 ug/kg Q1W
24 ug/kg Q2W
24 ug/kg Q3W
32 ug/kg Q1W
32 ug/kg Q2W
32 ug/kg Q3W
Fold Change
Time [days]

NK, natural killer; QW, every week; Q2W, every 2 weeks; Q3W, every 3 weeks.

Molecular response in ctDNA predicts clinical benefit in patients with advanced cancers treated with SAR'245 monotherapy

- Molecular response corresponded with Response Evaluation Criteria in Solid Tumors (RECIST) response and showed better best overall response and longer progression free survival

Figure 8: Pooled patients from cohort A, B, and G to evaluate association of ctDNA molecular response (a) within clinical response (b) with tumor size reduction

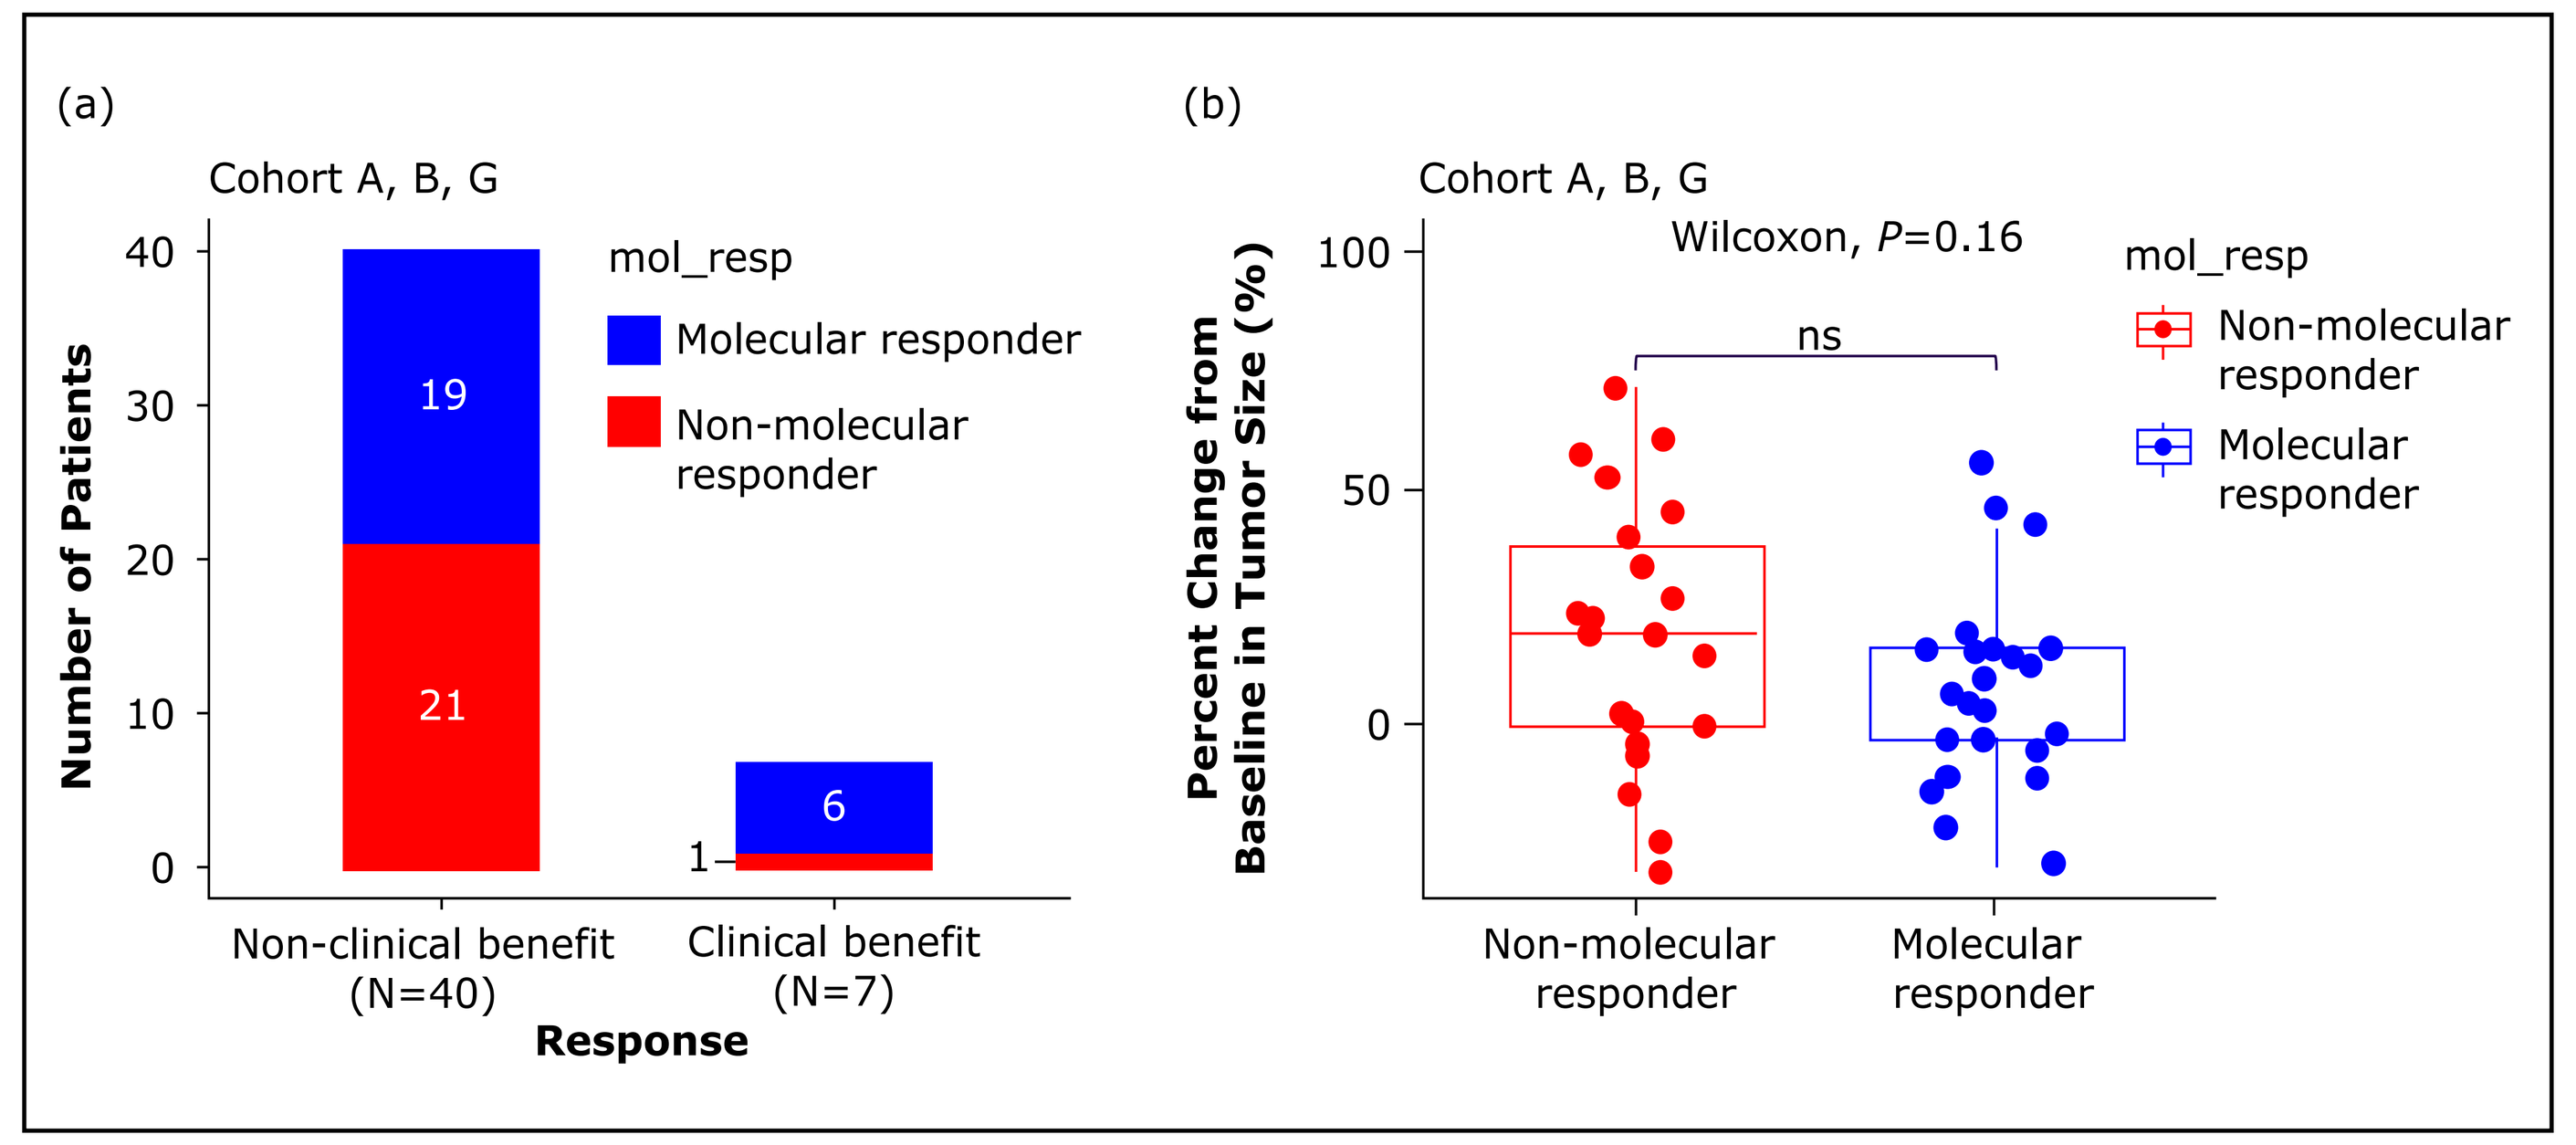
(a)
Cohort A, B, G
Number of Patients
mol_resp
Molecular responder
Non-molecular responder
19
21
6
1
Non-clinical benefit (N=40)
Clinical benefit (N=7)
Response
(b)
Cohort A, B, G
Wilcoxon, P=0.16
ns
Percent Change from Baseline in Tumor Size (%)
mol_resp
Non-molecular responder
Molecular responder
Non-molecular responder
Molecular responder

Efficacy surrogate biomarker circulating DNA (ctDNA) was measured from serum by Guardant Health. Clinical benefit (defined as partial or stable response lasting >6 months) vs. in non-clinical benefit (defined as progression of disease ≤6 months). ctDNA, circulating tumor DNA.

Figure 9. Molecular response showed longer PFS

Kaplan-Meier curve of PFS for patients treated with SAR'245 monotherapy stratified by ctDNA molecular responder and molecular non-responder

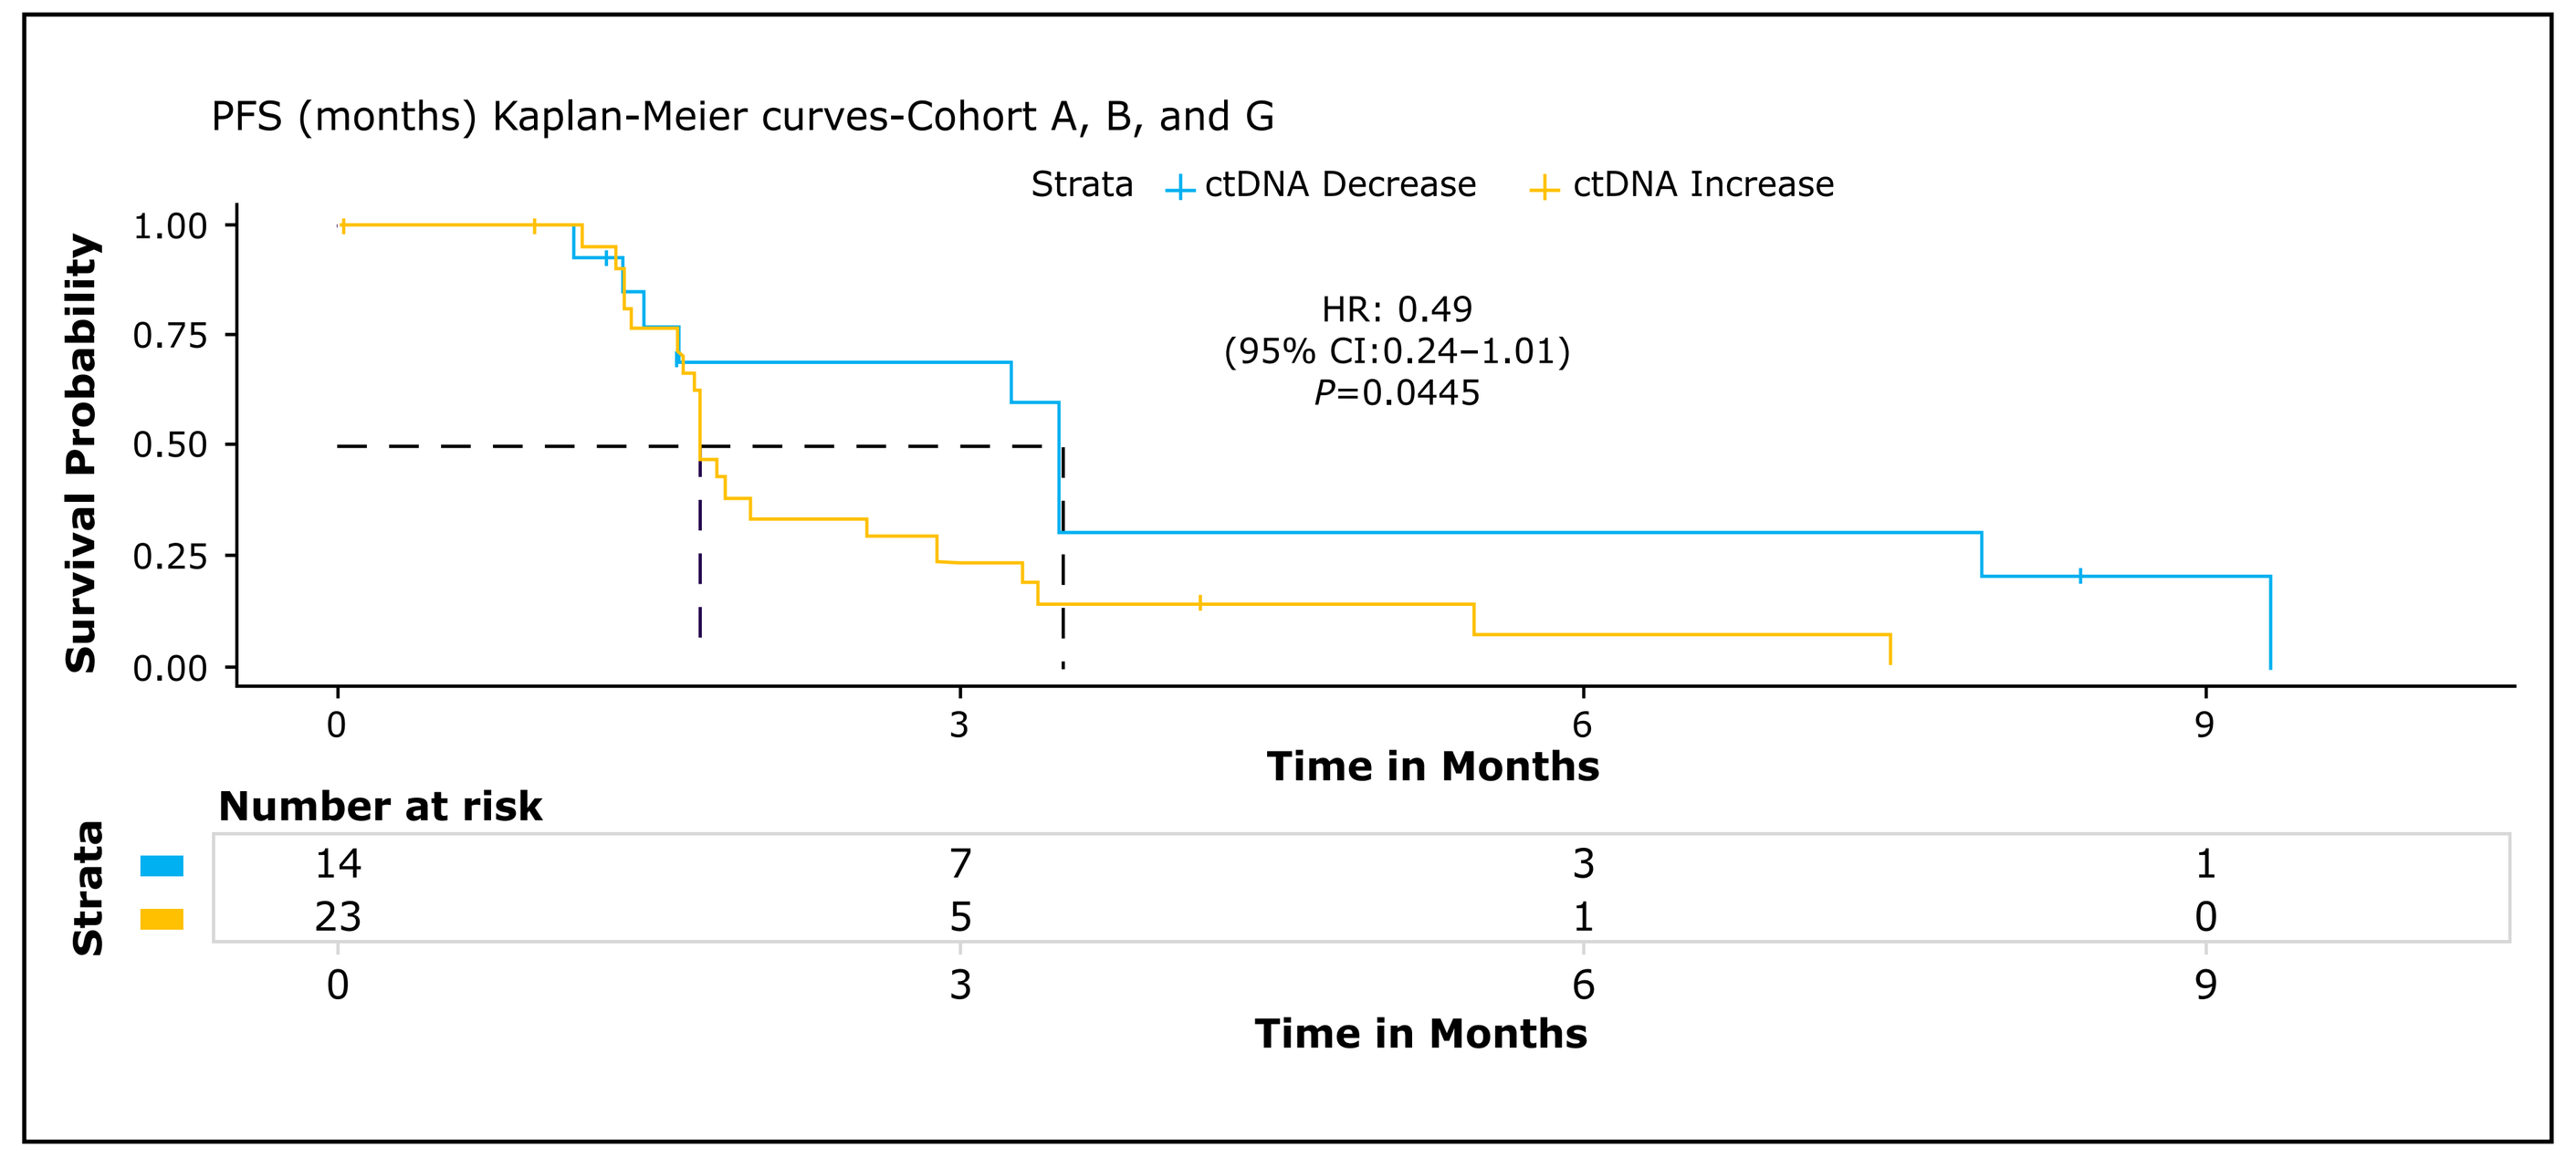
PFS (months) Kaplan-Meier curves-Cohort A, B, and G
Strata
ctDNA Decrease
ctDNA Increase
Survival Probability
HR: 0.49
(95% CI:0.24–1.01)
P=0.0445
Time in Months
Number at risk
Strata
14 7 3 1
23 5 1 0
0 3 6 9
Time in Months

Efficacy surrogate biomarker circulating DNA (ctDNA) was measured from serum by Guardant Health.
ctDNA, circulating tumor DNA; CI, confidence interval; HR, hazard ratio; PFS, progression free survival.

CONCLUSIONS

- We analyzed the HAMMER study of SAR'245 using a novel joint modeling platform that integrates response and MoA biomarkers as well as the PK/PD model in a simulation approach to support dose and schedule selection
- The study allows to overcome the limitations of small patient and biopsy numbers, and the intra- and inter-patient and tumor type heterogeneity common in early phase clinical studies
- The current PK/PD modeling results indicate that more frequent dosing of SAR'245 could lead to sustained expansion and activation of $CD8^+$ T cells and NK cells, while not triggering T_{reg} increase, and thus confirms the MoA and high selectivity to IL-2Rβγ complex

ACKNOWLEDGMENTS:

We acknowledge Tjokosela Tikiso from Sanofi, for contributions to PK/PD modeling. This study was sponsored by Sanofi. Medical writing support was provided by Aruna Meka, PhD and Deepa Warrier, PhD from Sanofi.

FUNDING:

This study is sponsored by Sanofi

DISCLOSURES:

Siqing Fu: Research Funding: Novartis, NeuPharma Inc, BeiGene, MacroGenics, BioAtla, Parexel International LLC, Boehringer Ingelheim, Abbisko, Lilly, Hookipa Pharma, IMV, Innovent Biologics, Lyvgen Biopharma, Millennium, Nerviano Medical Sciences, NIH/NCI, Sellas Life Sciences, Soricimed, NovoCure, Turnstone Bio, Taiho Oncology, NCCN, Exelis, Green2Bio Inc, K-Group Beta, NextCure, Ningbo NewBay Medical Technology, PureTech Health LLC, SQZ Biotech, Sumitomo Dainippon Pharma Oncology, Treadwell Therapeutics, Tyligand Bioscience Ltd, and Vaccibody. **Meredith Mckean:** Research Funding: Alpine Immune Sciences, Arcus Biosciences, Arvinas, Ascentage Pharma Group, Bayer, Bicycle Therapeutics, BioMed Valley Discoveries, BioNTech, Dragonfly Therapeutics, EMD Serono, Epizyme, Erasca, Exelixis, Foghorn Therapeutics, Genentech, Gilead Sciences, GlaxoSmithKline, IDEAYA Biosciences, Ikena Oncology, ImmVira Pharma, Infinity Pharmaceuticals, Jacobio Pharmaceuticals, Kechow Pharma, Kezar Life Sciences, Kinnate BioPharma, MedImmune, Mereo BioPharma, Metabomed, Moderna, NBE Therapeutics, Nektar, Novartis, Oncorus, PACT Pharma, Pfizer, Plexxikon, Prelude Therapeutics, Pyramid Biosciences, Regeneron, Sapience Therapeutics, Scholar Rock, Seattle Genetics, Synthrox, Takeda Pharmaceuticals, Teneobio, Tempest Therapeutics, Tizona Therapeutics, TMUNITY Therapeutics, TopAlliance Biosciences, and Xilio; and Consulting/Advisory role: Astellas Pharma, AstraZeneca, BicycleTX Limited, Castle Biosciences, Eisai, Ideaya Biosciences, iTeos, Moderna, Pfizer, and Regeneron Pharmaceuticals. **Gerald Falchook:** Research funding: 3-V Biosciences, Abbisko, Abbvie, ABL Bio, ADC Therapeutics, Accutar, Aileron, American Society of Clinical Oncology, Amgen, ARMO/Eli Lilly, Artios, AstraZeneca, BeiGene, Bioatla, Bioinvent, Biothera, Bicycle, Black Diamond, Boehringer Ingelheim, Celldex, Celgene, Ciclomed, Curegenix, Curis, Cyteir, Daiichi, DelMar, eFFECTOR, Eli Lilly, EMD Serono, Epizyme, Erasca, Exelixis, Freenome, Fujifilm, Genmab, GlaxoSmithKline, Hutchison MediPharma, IGM Biosciences, Ignyta, ImmunoGen/MacroGenics, Incyte, Jacobio, Jounce, Jubilant, Kolltan, Loxo/Bayer, MedImmune, Millennium, Merck, miRNA Therapeutics, Molecular Templates, National Institutes of Health, Navire, NiKang, Novartis, OncoMed, Oncorus, Oncothyreon, Poseida, Precision Oncology, Prelude, PureTech, Pyramid, RasCal, Regeneron, Relay, Rgenix, Ribon, Samumed, Sapience, Seagen, Silicon, Simcha, Sirnaomics, Strategia, Syndax, Synthorx/Sanofi, Taiho, Takeda, Tarveda, Teneobio, Tesaro, Tocagen, Turning Point, U.T. MD Anderson Cancer Center, Vegenics, Xencor; Advisory role: Fujifilm, Silicon, Navire, Turning Point, Predicine, Inspirna, Regeneron; Advisor: EMD Serono (2010, 2011); Honorarium: Total Health Conferencing (2019), Rocky Mountain Oncology Society (2020); and Travel (self, for work and/or research related to institution): BMS, EMD Serono, Fujifilm, Millennium, and Sarah Cannon Research Institute (employer, annually). **Minal Barve:** Research Funding: Mary Crowley Cancer Research; Stock Ownership: Texas Oncology Physician Associates. **Cheng Ean Chee:** Consulting/Advisory Role: Guardant Health AMEA; AstraZeneca; Roche/Genentech, Pierre Fabre, Merck. **Charlotte Lemech:** None. **Tira J. Tan:** Financial Interests (personal): Astra Zeneca, Advisory Board; MSD, Advisory Board; Astra Zeneca, Invited Speaker; Novartis, Invited Speaker; MSD, Invited Speaker; Gilead, Invited Speaker; Daiichi Sankyo, Advisory Board; Gilead, Advisory Board; Novartis, Advisory Board; Astra Zeneca, Research Grant. Financial interest (institutional): Roche, Local PI; Novartis, Local PI; Astra Zeneca, Local PI; Daiichi Sankyo, Local PI; Genentech, Local PI; Sanofi, Local PI; Loxo, Local PI; Gilead, Local PI. Non-Financial Interests: ASCO, Member. Other (Conference sponsorship): Astra Zeneca, Pfizer, and MSD. **Tarek Meniawy:** Honoraria: AstraZeneca. **Neyssa Marina:** Currently employed by Arcus Biosciences (employed by Sanofi at the time of the study). **Emiliano Calvo Aller:** Advisory board participation (financial interest) for Adcendo, Alkermes, Amunix, Anaveon, Amcure, AstraZeneca, BMS, Janssen, MonTa, MSD, Nanobiotix, Nouscom, Novartis, OncoDNA, PharmaMar, Roche/Genentech, Sanofi, Servier, SyneosHealth, TargImmune, and T-knife; research grants for Achilles and BeiGene; steering committee (financial interest) and IDMC participation for BeiGene (IDMC steering committee), EORTC IDMC chair (non-financial interest), MedSIR (steering committee), and Novartis (steering committee); honoraria for scientific board participation for Adcendo, Chugai Pharmaceuticals, and PsiOxus Therapeutics; employment at HM Hospitals Group and START Program of Early Phase Clinical Drug Development in Oncology, START corporation, Oncoart Associated, and International Cancer Consultants and is the founder and president of the not-for-profit Investigational Therapeutics in Oncological Sciences (INTHEOS) Foundation. **Giovanni Abbadessa:** Currently employed by ModeX Therapeutics Inc. (employed by Sanofi at the time of the study). **Wenting Wang, Adyb Baakili, Silke Gastine, and Rui Wang** Employed by Sanofi.

REFERENCES:

1. Falchook G, et al. *BMJ Specialist Journals*.2021;P481:9.
2. Fu, Siqing, et al. *Cancer Research* 83.7_Supplement (2023): 2155-2155.
3. NCT04009681 available from website ClinicalTrials.gov.
4. Conlon KC, et al. *Journal of Interferon & Cytokine Research*. 2019;39(1):6-21.
5. Diab A, et al. *Journal of Clinical Oncology*. 2023;41(30):4756-4767.
6. Tannir NM, et al. *Journal of Clinical Oncology*. 2024;42(23):2800-2811.

QR CODE:

Copies of this poster obtained through Quick Response (QR) Code are for personal use only
For questions, please contact
Rui Wang at rui.wang@sanofi.com

Society for Immunotherapy of Cancer (SITC) Annual Meeting 2024 – Houston, TX, Nov 6-10, 2024